#2930: Synergistic and additive anti-tumor effects of MIV-818 in combination with sorafenib in nonclinical hepatocellular carcinoma models

Biljana Rizoska[1], Johan Bylund[1], Sveinn Briem[1], Alastair Kyle[2], Andrew Minchinton[2], Fredrik Öberg[1], Karin Göhlin[1] and Mark Albertella[1]
[1]Medivir AB, Huddinge, Sweden; [2]Cabenda, Vancouver, BC Cancer Research Center, BC, Canada

MEDIVIR

Contact person: Mark.Albertella@medivir.com

Background

- **MIV-818, a nucleotide prodrug of troxacitabine-monophosphate, has been designed to target the chain-terminating nucleotide troxacitabine-triphosphate (TRX-TP) to the liver after oral dosing, while minimizing systemic exposure**
- **Sorafenib is a multikinase inhibitor with antiangiogenic and antiproliferative effects that is approved for the first-line treatment of advanced hepatocellular carcinoma (HCC)**
- **Hypoxia is induced as a result of the antiangiogenic effects of sorafenib. Since hypoxic conditions have been shown to increase cytotoxicity of TRX via increased conversion of TRX-diphosphate to TRX-TP, we investigated the effects of combining MIV-818 or TRX with sorafenib in cell lines and in xenograft mouse models of HCC**

Methods

In vivo rat PK
MIV-818 (80 µmol/kg, PO) and TRX (80 µmol/kg, PO and IV) were administered to male Wistar rats and plasma and liver were collected at different time points after dosing (up to 24 h) for bioanalysis.

In vivo xenograft models
HCC subcutaneous xenograft models were established by inoculation of Hep3B (2x10^6) or HepG2 (1x10^7) cells (0.1 mL in 1:1 PBS:Matrigel) subcutaneously into the left flank of Balb/C nude female mice. Treatment was initiated when a tumour volume of ~200 mm^3 was reached. TRX was dosed intraperitoneally (ip) BID for 5 days at 2.5, 10 and 25 mg/kg. MIV-818 was dosed via oral gavage BID for 5 days at 48 and 160 µmol/kg. Sorafenib was dosed at 30 mg/kg PO QD for 21 days. Tumours were measured using electronic callipers and volumes were estimated using the formula 0.5 (LxW^2). For efficacy studies the animals were monitored until a terminal size tumour was reached or until a relative tumour volume (RTV) times four the initial TV at start of treatment was reached. For the PD studies the mice were injected ip with a BrdU/pimonidazole (600mg/kg/60mg/kg) mixture 2 h prior to being terminated at 2 h after the last dose. Tumour was collected for bioanalysis and histology.

Histology
Tumour cryosections (10 µm) were immunostained for hypoxia using mouse-anti-pimonidazole-FITC (1:500), anti-phospho-Histone H2A.X (Ser139) mouse-anti-human-gH2AX (Clone JBW301) tagged with Alexa 647, BrdU using a monoclonal rat-anti-BrdU (clone BU/175; 1:500) and anti-mouse Alexa 750 secondary (1:500). Cellular DNA was counter-stained with Hoechst 33342. The imaging system consisted of a robotic fluorescence microscope with a PCO Edge 4.2 camera and customized ImageJ software. Images of BrdU, pH2AX, pimonidazole & Hoechst 33342 staining from each tumour section were overlaid and areas of necrosis, acellular cavities and staining artifacts manually removed. Positive regions for each marker were identified by selecting all pixels above tissue background levels. Analysis of whole tissue averages for each marker were determined by dividing the total number of positive pixels by the total tissue area excluding necrosis and empty regions.

Bioanalysis
Determination of TRX in plasma and TRX-TP concentrations in liver or tumour homogenates was performed using LC-MS/MS.

MIV-818: In vitro properties

- **MIV-818 demonstrates pronounced anti-proliferative activity against HCC cell lines, with 3 to 5-fold greater potency than troxacitabine**
- **MIV-818 induces a strong increase in DNA damage in the HepG2 cell line (56-fold), while a minor induction (2-fold) is seen in primary human hepatocytes**
- **MIV-818 does not affect the viability of primary human hepatocytes**
- **Cytotoxic effects are dependent on cells undergoing division which also creates a potential therapeutic index preferentially targeting cancer cells versus normal cells due to the differences in proliferation rate**
- **MIV-818 is stable in human, dog and cynomologus monkey whole blood although unstable in rodent blood**
- **The rapid hydrolysis of MIV-818 in mouse blood limits the possibility to evaluate efficacy of MIV-818 in mouse models of disease**

Assay	MIV-818	Troxacitabine
Hep3B growth inhibition, mean IC_{50}	0.15 µM	0.40 µM
HepG2 growth inhibition, mean IC_{50}	0.019 µM	0.092 µM
HepG2 DNA damage (p-p53)	0.033 µM (56-fold)	-
Primary human hepatocytes – viability IC_{50}	>100 µM	>100 µM
Primary human hepatocytes - DNA damage (p-p53)	no value (2-fold)	-
Human, dog, monkey whole blood CL_{int} (µL/min/mg)	Stable (<2)	Stable (<2)
Mouse, rat whole blood CL_{int} (µL/min/mg)	Very unstable (>150)	Stable (<2)

MIV-818 liver targeting in rat

- **Troxacitabine (TRX) at 80 µmol/kg was dosed IV or PO and MIV-818 at 80 µmol/kg was dosed PO to rats. The exposures to TRX in plasma and TRX-TP in liver were assessed.**

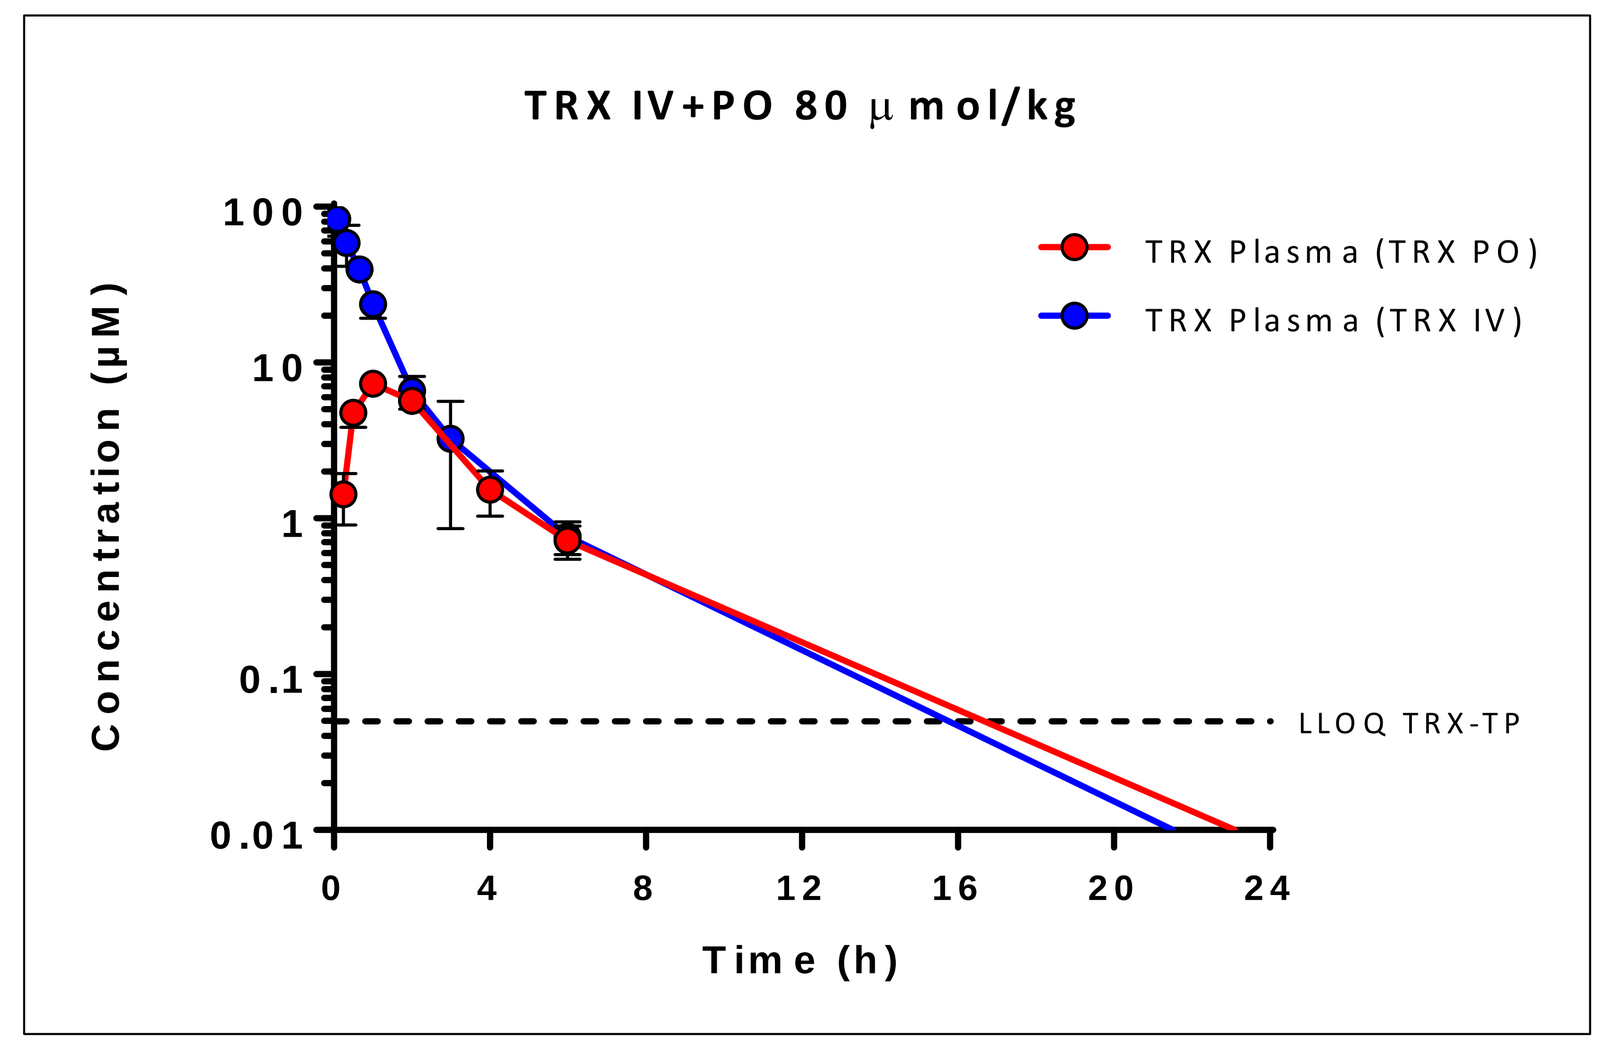
TRX IV+PO 80 µmol/kg
TRX Plasma (TRX PO)
TRX Plasma (TRX IV)
LLOQ TRX-TP
Concentration (µM)
Time (h)

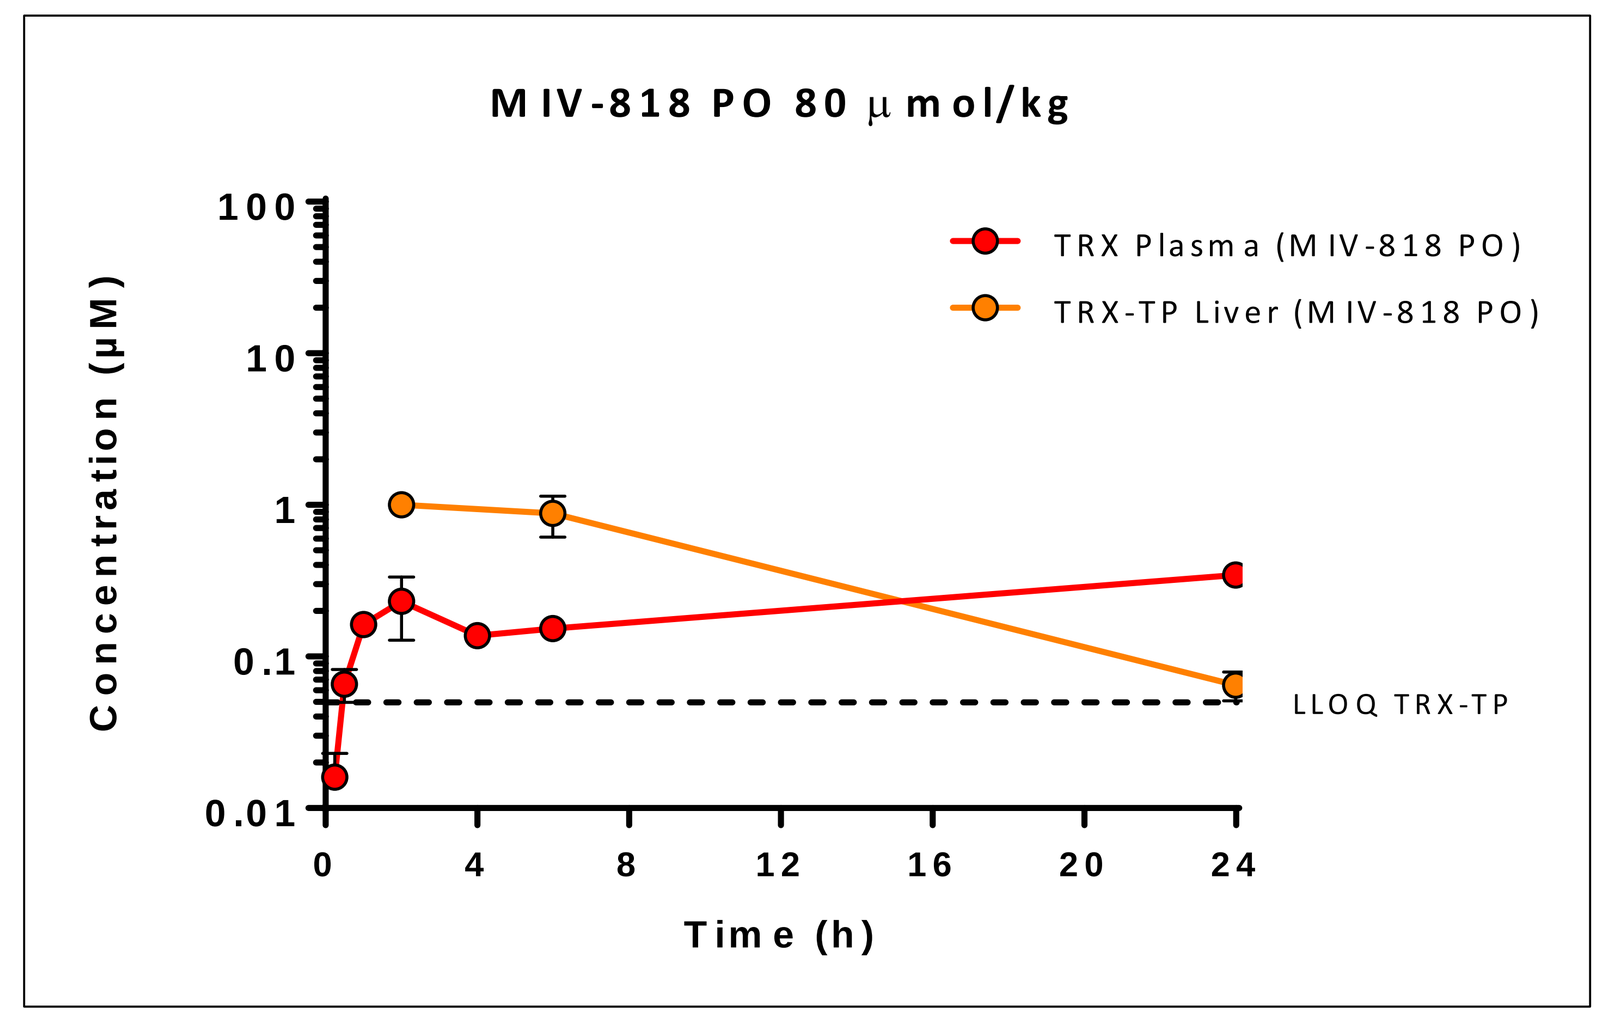
MIV-818 PO 80 µmol/kg
TRX Plasma (MIV-818 PO)
TRX-TP Liver (MIV-818 PO)
LLOQ TRX-TP
Concentration (µM)
Time (h)

Drug	Dose (µmol/kg)	Route	Analyte	Tissue	C_{max} (µM)	AUC_{0-24h} (µM*h)	TP_{liver} / TRX_{plasma} ratio
TRX	80	IV	TRX TRX-TP	Plasma Liver	102 <0.05	76 <1.2	**<0.016**
TRX	80	PO	TRX TRX-TP	Plasma Liver	7.4 <0.05	22 <1.2	**<0.055**
MIV-818	80	PO	TRX TRX-TP	Plasma Liver	0.35 1.0	5.4 10	**1.9**

- **MIV-818 administered PO resulted in a liver TRX-TP Cmax of 1.0 µM and a liver TRX-TP AUC_{0-24} of 10 µM*h**
- **The same dose of TRX dosed IV and PO resulted in liver TRX-TP concentrations below the limit of quantification (0.05 µM) at all time points**
- **Cmax liver TRX-TP was >20 times higher for MIV-818 after PO dosing than for TRX after IV and PO dosing**
- **AUC_{0-24} liver TRX-TP vs. AUC_{0-24} plasma TRX ratio for MIV-818 after PO dosing was >100 times higher than for TRX after IV dosing, demonstrating the substantially improved liver targeting by MIV-818 in rat, despite low stability in rat blood**

MIV-818 synergy with sorafenib in vitro

- **MIV-818 shows synergy in vitro with sorafenib in Hep3B cells**

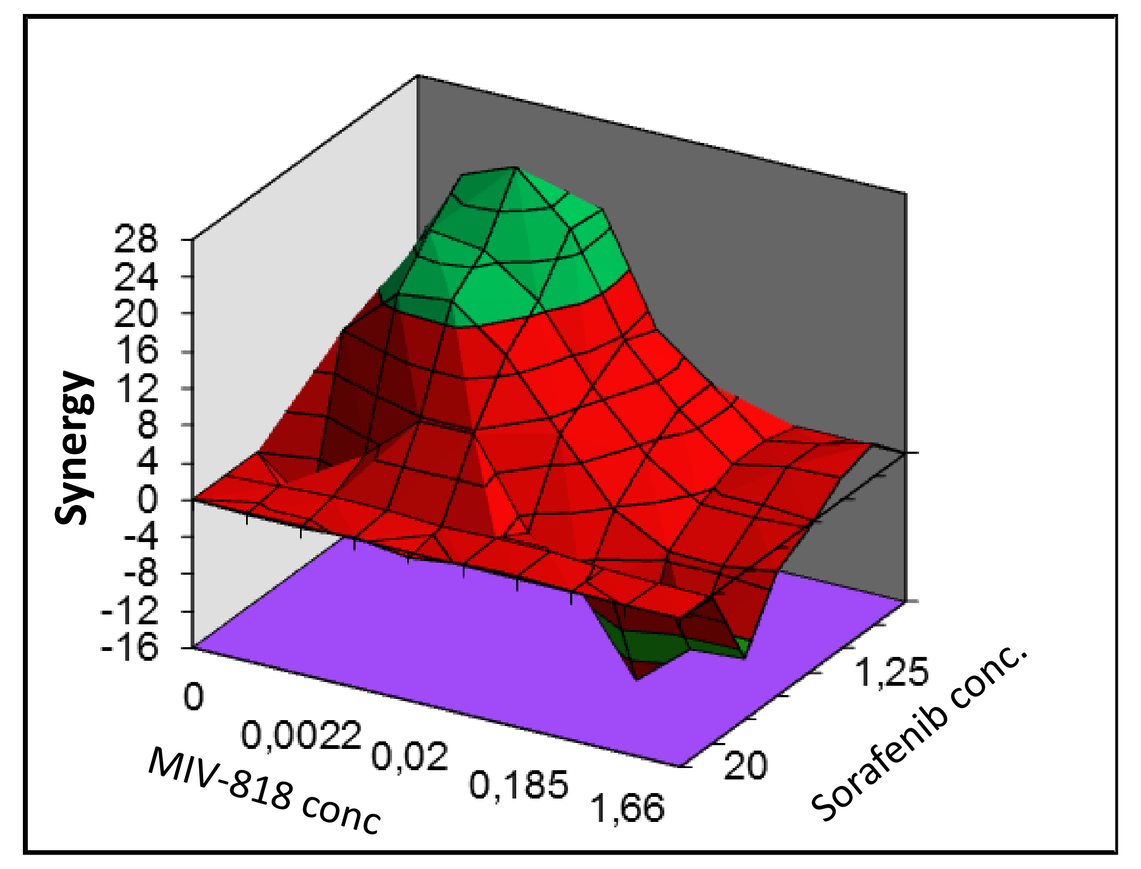
Synergy
MIV-818 conc
Sorafenib conc.

Hep3B proliferation
WST-8 assay

Synergy plot generated using MacSynergy II software. Hills indicate synergistic interactions.

Tumour growth inhibition after combination of troxacitabine and sorafenib

- **Dose-dependent anti-tumour effects were observed after dosing with TRX at 2.5, 10 and 25 mg/kg (IP) twice daily for 5 days in the Hep3B model (A)**
- **Additive anti-tumour effects were observed after dosing with TRX at 2.5 mg/kg (IP) twice daily for 5 days and sorafenib at 30 mg/kg (PO) once daily for 21 days in the Hep3B model (B)**
- **Exposures of TRX and sorafenib in plasma and tumor were similar in the combination and single agent groups, suggesting no pharmacokinetic interactions**

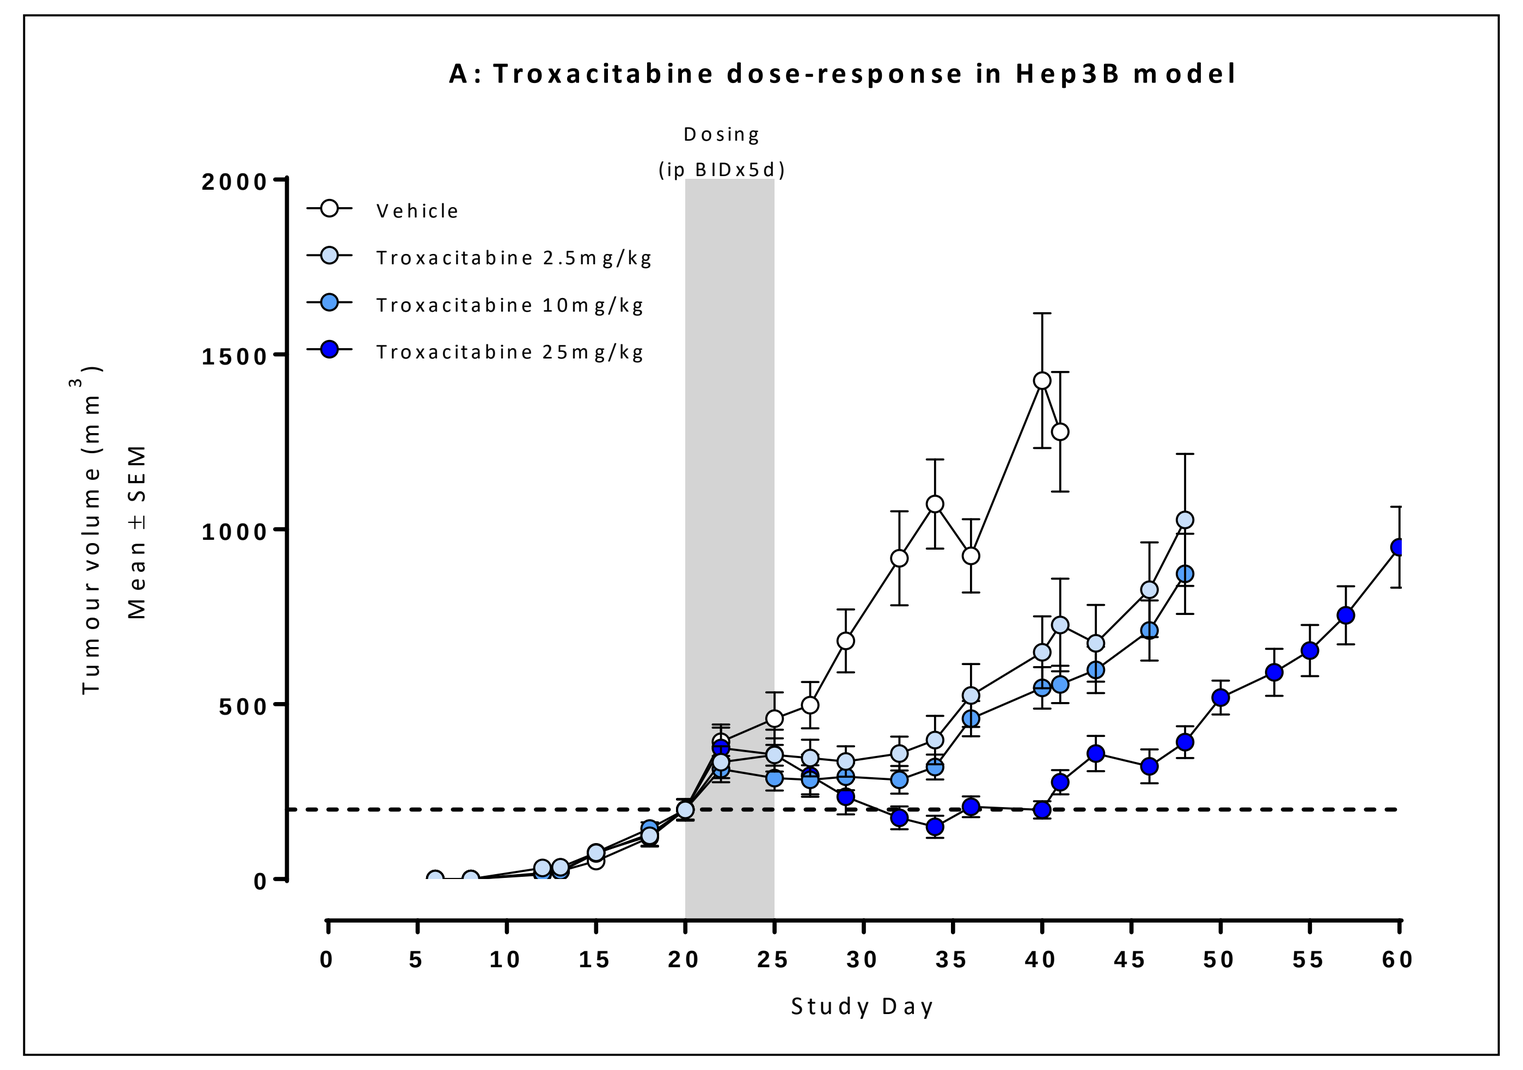
A: Troxacitabine dose-response in Hep3B model
Dosing (ip BIDx5d)
Vehicle
Troxacitabine 2.5mg/kg
Troxacitabine 10mg/kg
Troxacitabine 25mg/kg
Tumour volume (mm³) Mean ± SEM
Study Day

Group / dose	TGI (day 34)	TGD
Troxacitabine 2.5 mg/kg	70%	11d
Troxacitabine 10 mg/kg	81%	16d
Troxacitabine 25 mg/kg	101%	26d

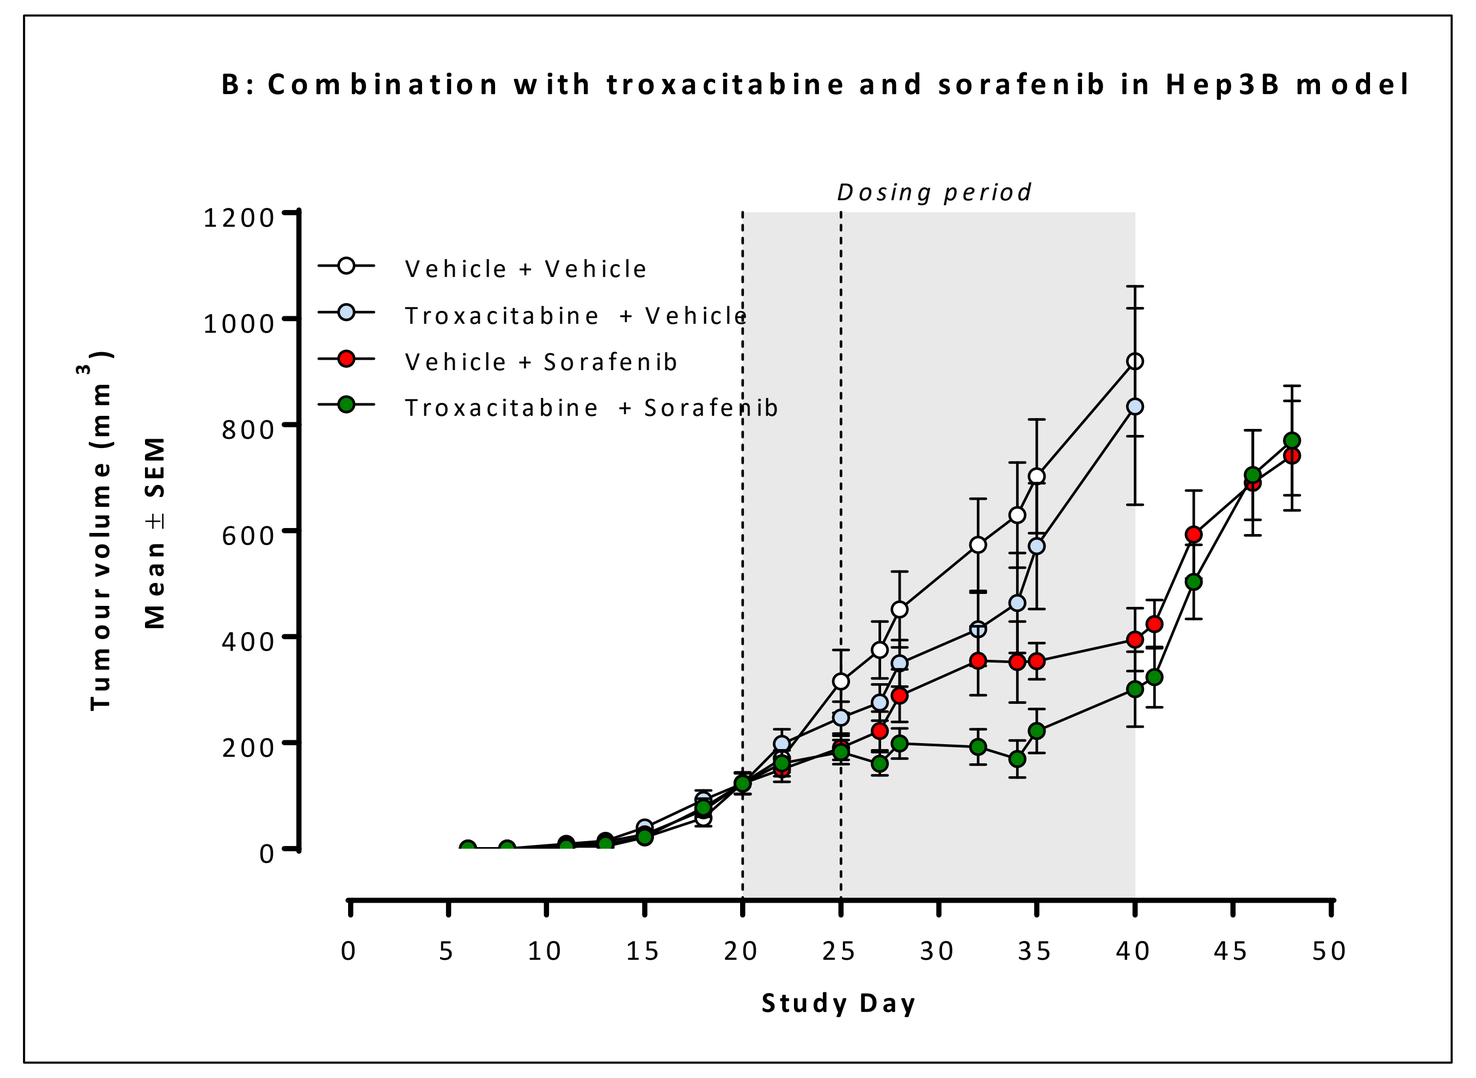
B: Combination with troxacitabine and sorafenib in Hep3B model
Dosing period
Vehicle + Vehicle
Troxacitabine + Vehicle
Vehicle + Sorafenib
Troxacitabine + Sorafenib
Tumour volume (mm³) Mean ± SEM
Study Day

Group / dose	TGI (day 34)	TGD
Troxacitabine 2.5 mg/kg	32%	5.5
Sorafenib 30 mg/kg	52%	11
Troxacitabine + sorafenib	90%	13.5

Tumour growth inhibition after combination of MIV-818 and sorafenib

- **Dose-dependent anti-tumor effects were observed after dosing with MIV-818 at 48 and 160 µmol/kg (PO) twice daily for 5 days in the HepG2 model (A)**
- **Additive anti-tumor effects were observed after dosing with MIV-818 at 160 µmol/kg (PO) twice daily for 5 days and sorafenib at 30 mg/kg (PO) once daily for 21 days in the HepG2 model (B)**
- **Note that sorafenib was only marginally effective in this model**

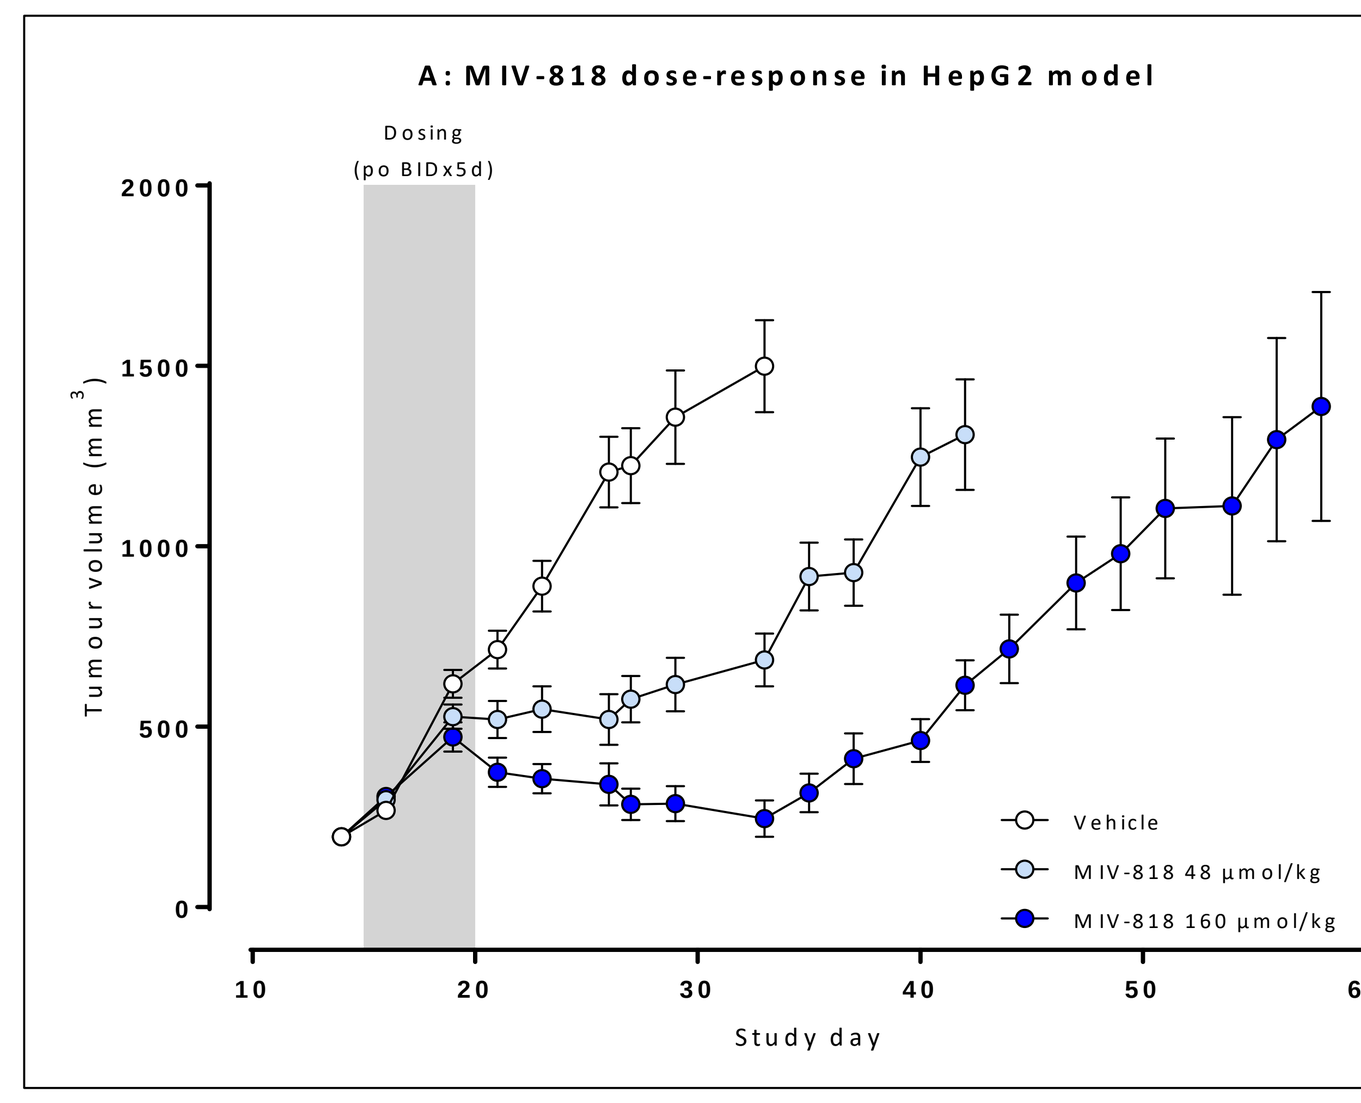
A: MIV-818 dose-response in HepG2 model
Dosing (po BIDx5d)
Vehicle
MIV-818 48 µmol/kg
MIV-818 160 µmol/kg
Tumour volume (mm³)
Study day

Group / dose	TGI (day 33)	TGD
MIV-818 48 µmol/kg	63%	12d
MIV-818 160 µmol/kg	96%	23d

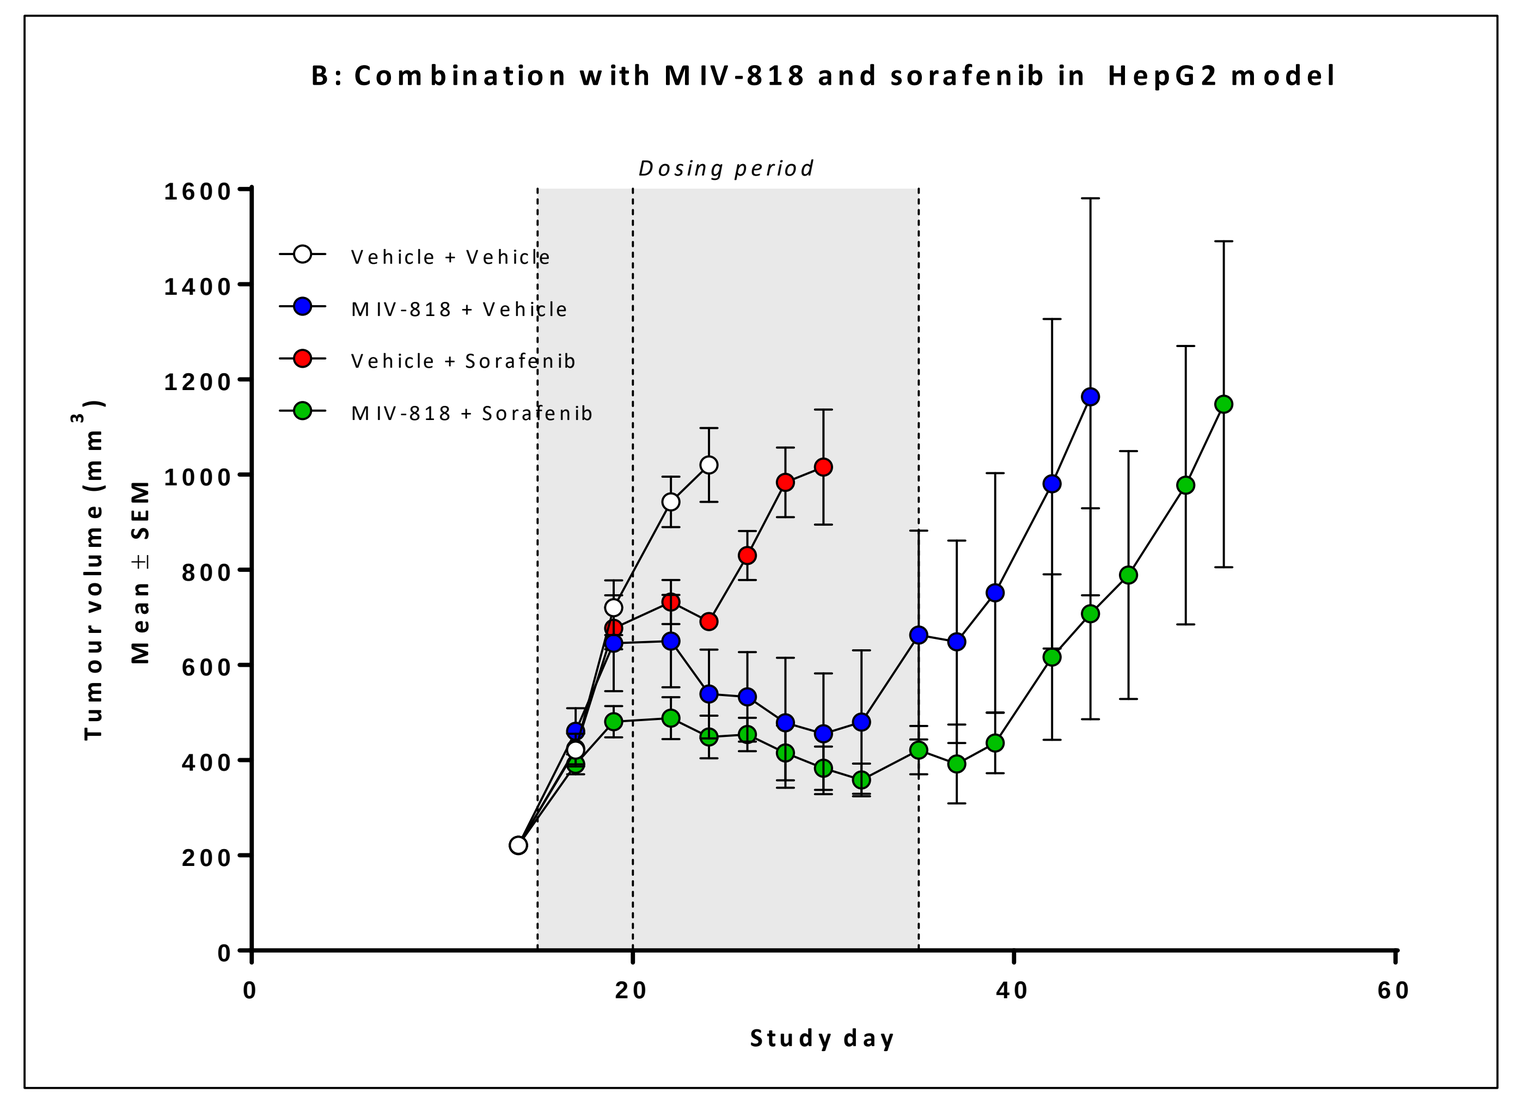
B: Combination with MIV-818 and sorafenib in HepG2 model
Dosing period
Vehicle + Vehicle
MIV-818 + Vehicle
Vehicle + Sorafenib
MIV-818 + Sorafenib
Tumour volume (mm³) Mean ± SEM
Study day

Group / dose	TGI (day 24)	TGD
MIV-818 160 µmol/kg	63%	22d
Sorafenib 30mg/kg	39%	6d
MIV-818 + sorafenib	72%	26d

PD effects of troxacitabine and sorafenib

- **Substantial increase in hypoxia (pimonidazole) was seen in the sorafenib treated groups only (app. 3-fold vs. vehicle), consistent with the mode of action**
- **Substantial inhibition of proliferation (BrdU) in animals receiving TRX alone (by 80% vs. vehicle) or in combination with sorafenib (by 92% vs. vehicle), while a small, but significant, inhibition of proliferation was observed with sorafenib alone (by 27% vs. vehicle)**
- **Substantial induction of DNA damage (pH2AX) in animals receiving TRX alone or in combination with sorafenib (15-fold vs. vehicle), while no induction in the sorafenib alone group**
- **Induction of DNA damage, and reduced proliferation was also evident in hypoxic regions**

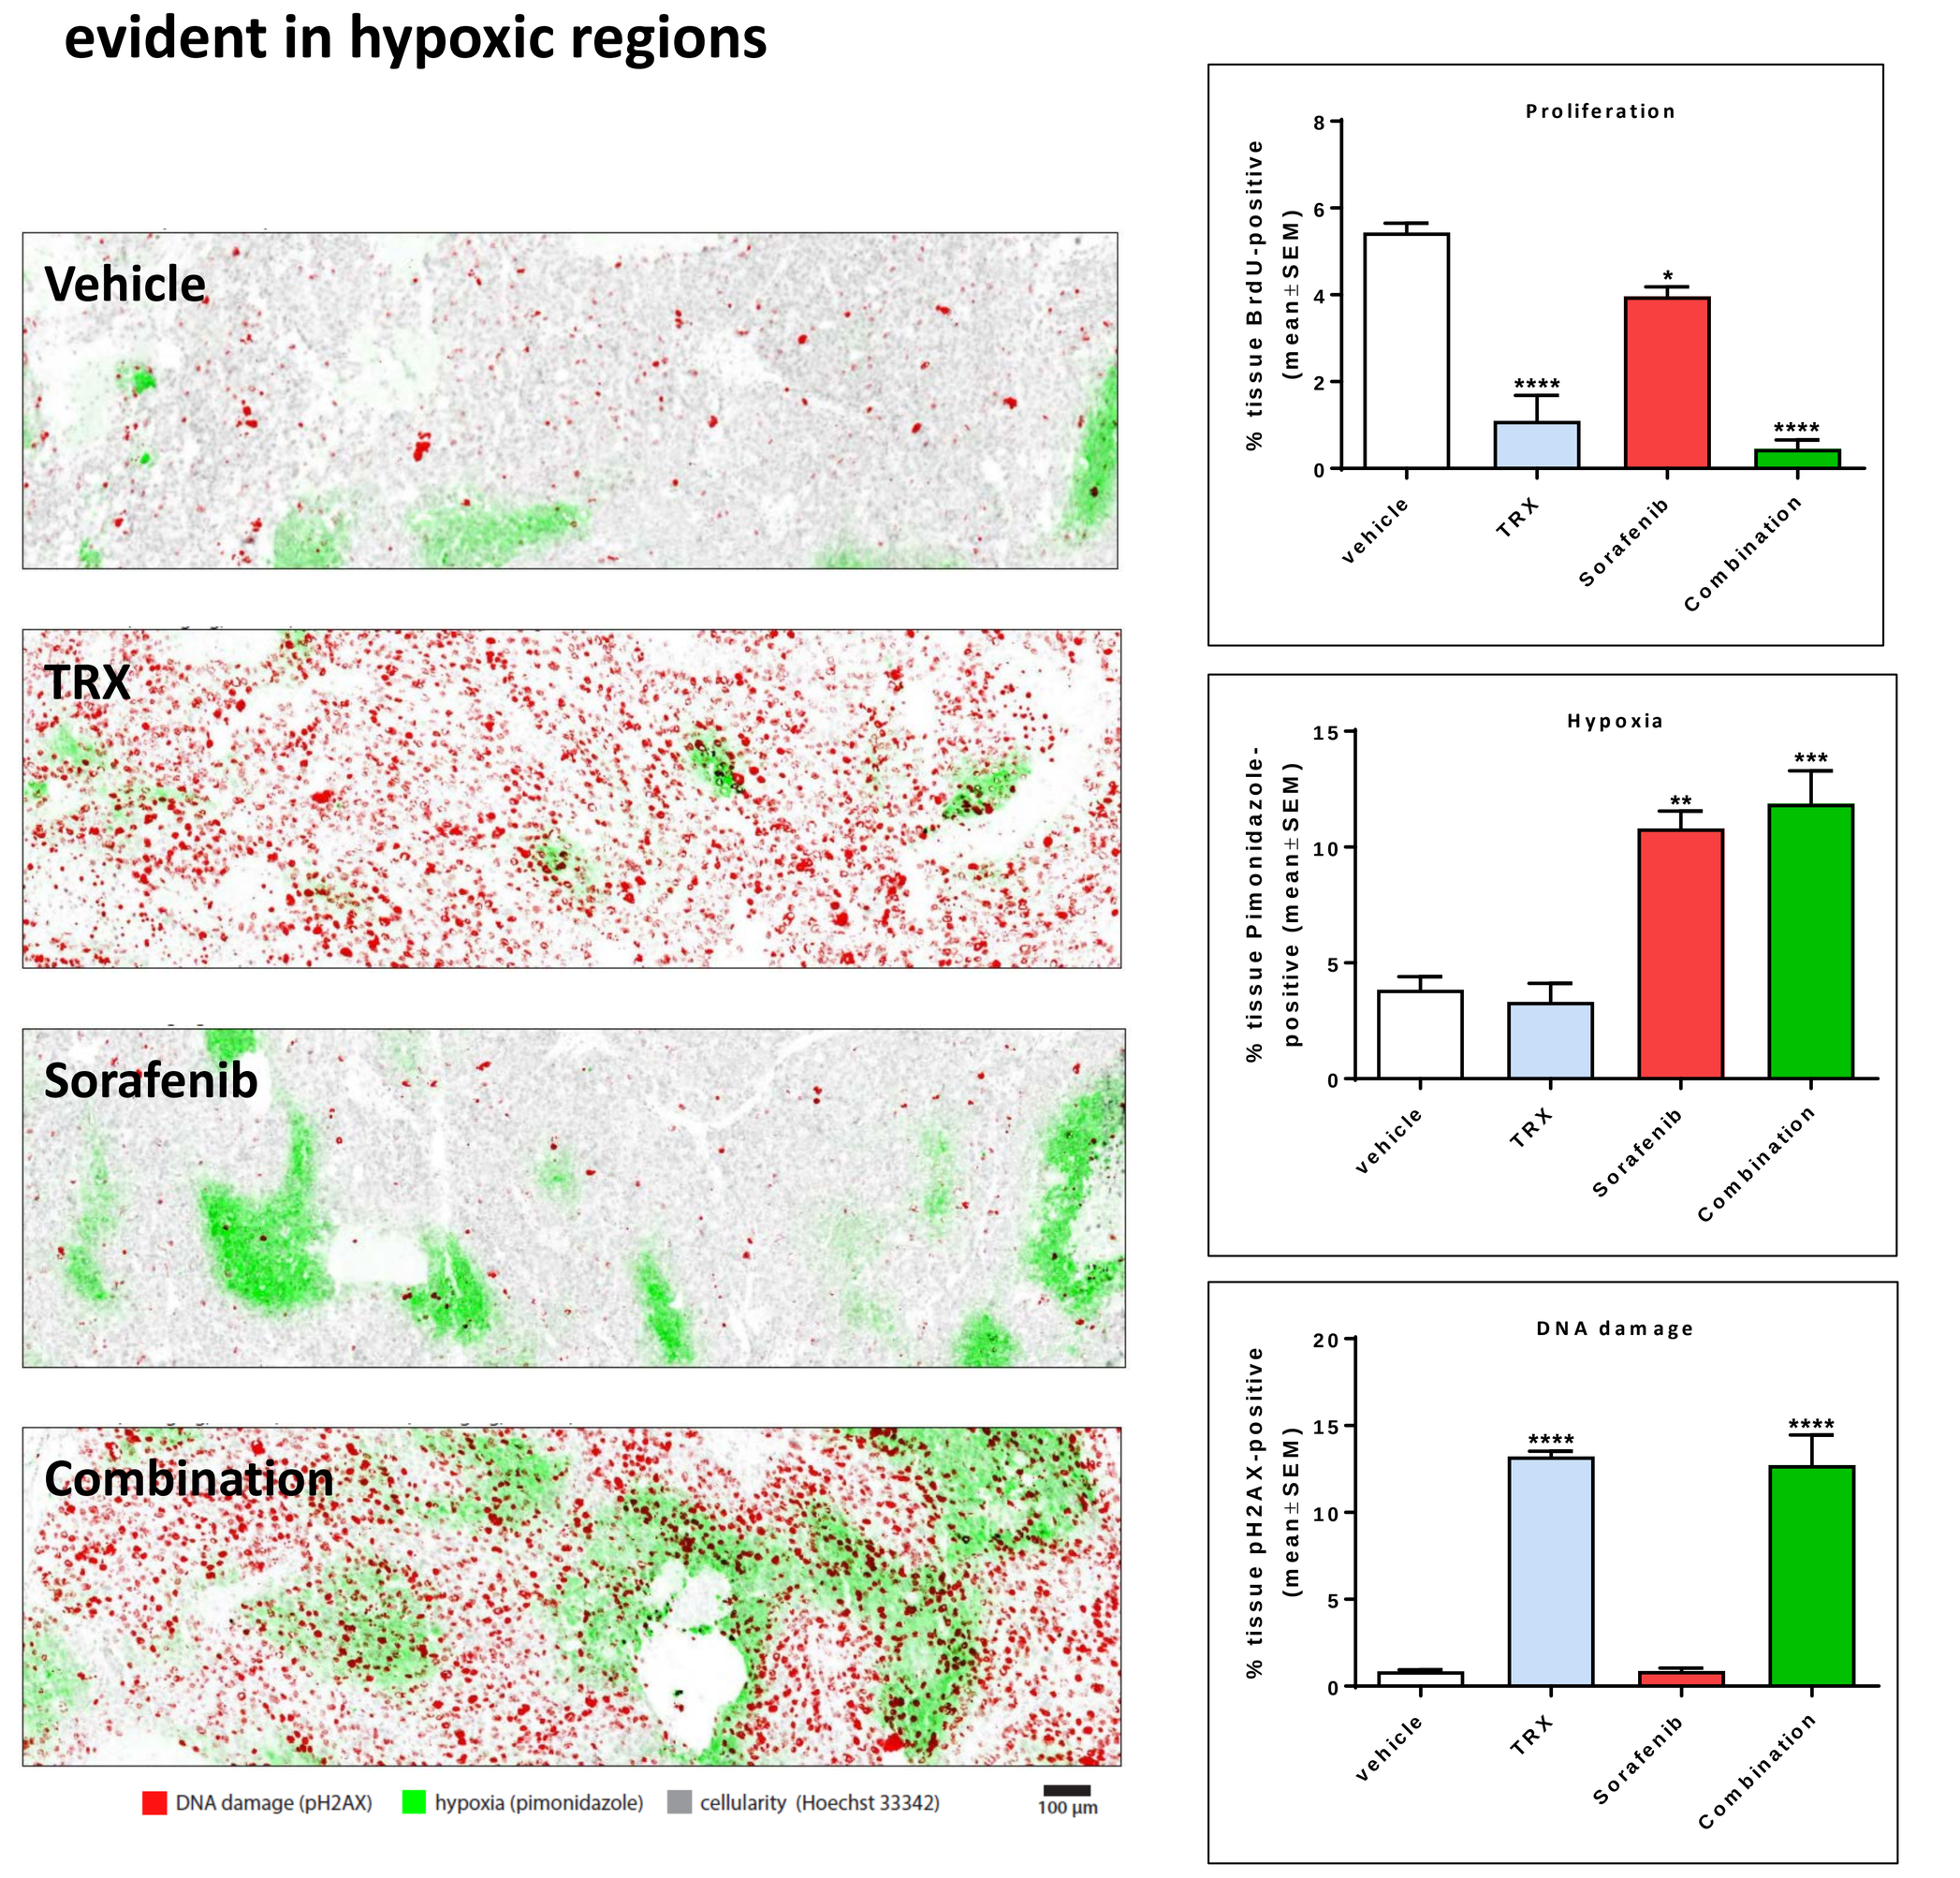
Vehicle
TRX
Sorafenib
Combination
DNA damage (pH2AX)
hypoxia (pimonidazole)
cellularity (Hoechst 33342)
100 µm
Proliferation
Hypoxia
DNA damage
% tissue BrdU-positive (mean±SEM)
% tissue Pimonidazole-positive (mean±SEM)
% tissue pH2AX-positive (mean±SEM)

*P<0.05, **P<0.01, *** P<0.001, ****P<0.0001 vs. vehicle

PD effects of MIV-818 and sorafenib

- **Combination of MIV-818 and sorafenib showed the same PD effects as for TRX (see below)**
- **Substantial inhibition of proliferation and induction of DNA damage**

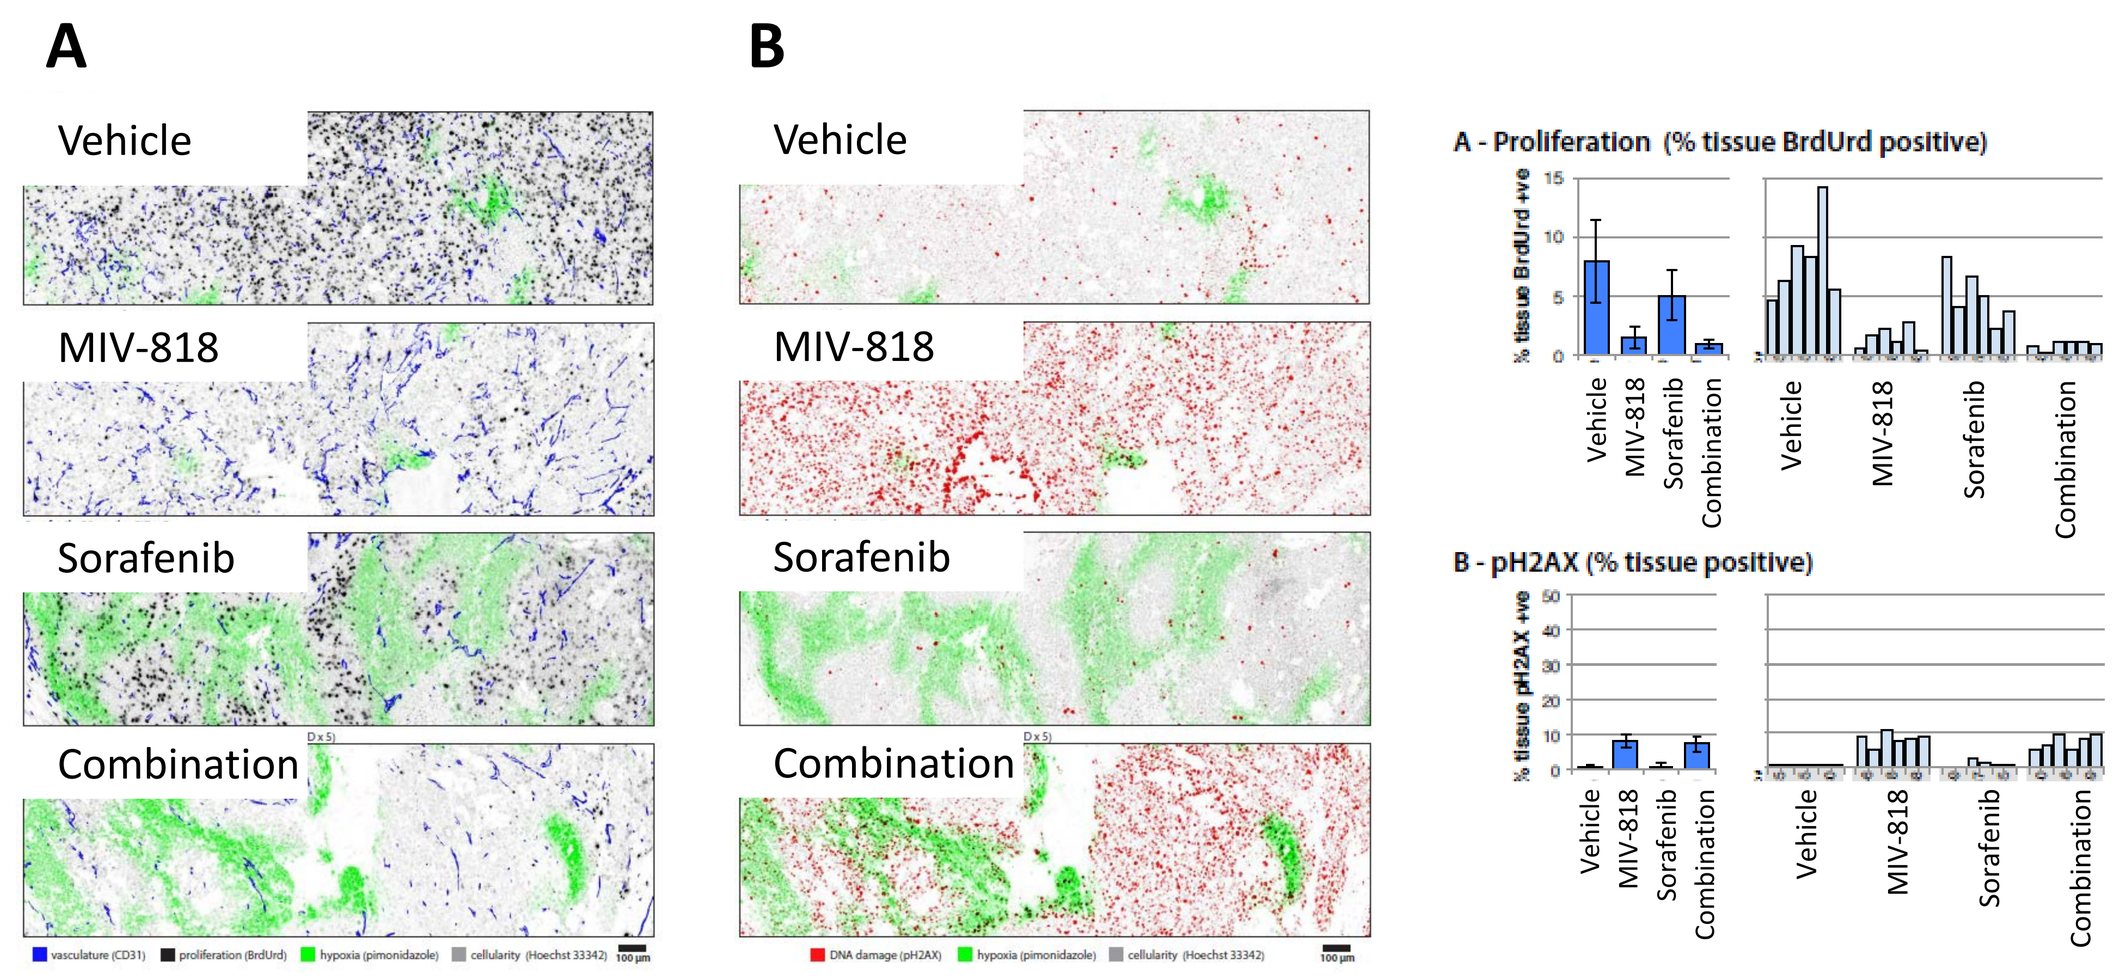
A
B
Vehicle
MIV-818
Sorafenib
Combination
A - Proliferation (% tissue BrdUrd positive)
B - pH2AX (% tissue positive)

Conclusions

- **MIV-818 and sorafenib are synergistic in vitro and the combination shows enhanced activity in vivo compared to either agent alone.**
- **The results suggest that add-on of MIV-818 to sorafenib may be beneficial for the treatment of HCC**
- **MIV-818 is currently in preparation for clinical trials in patients with advanced HCC and other liver cancers**